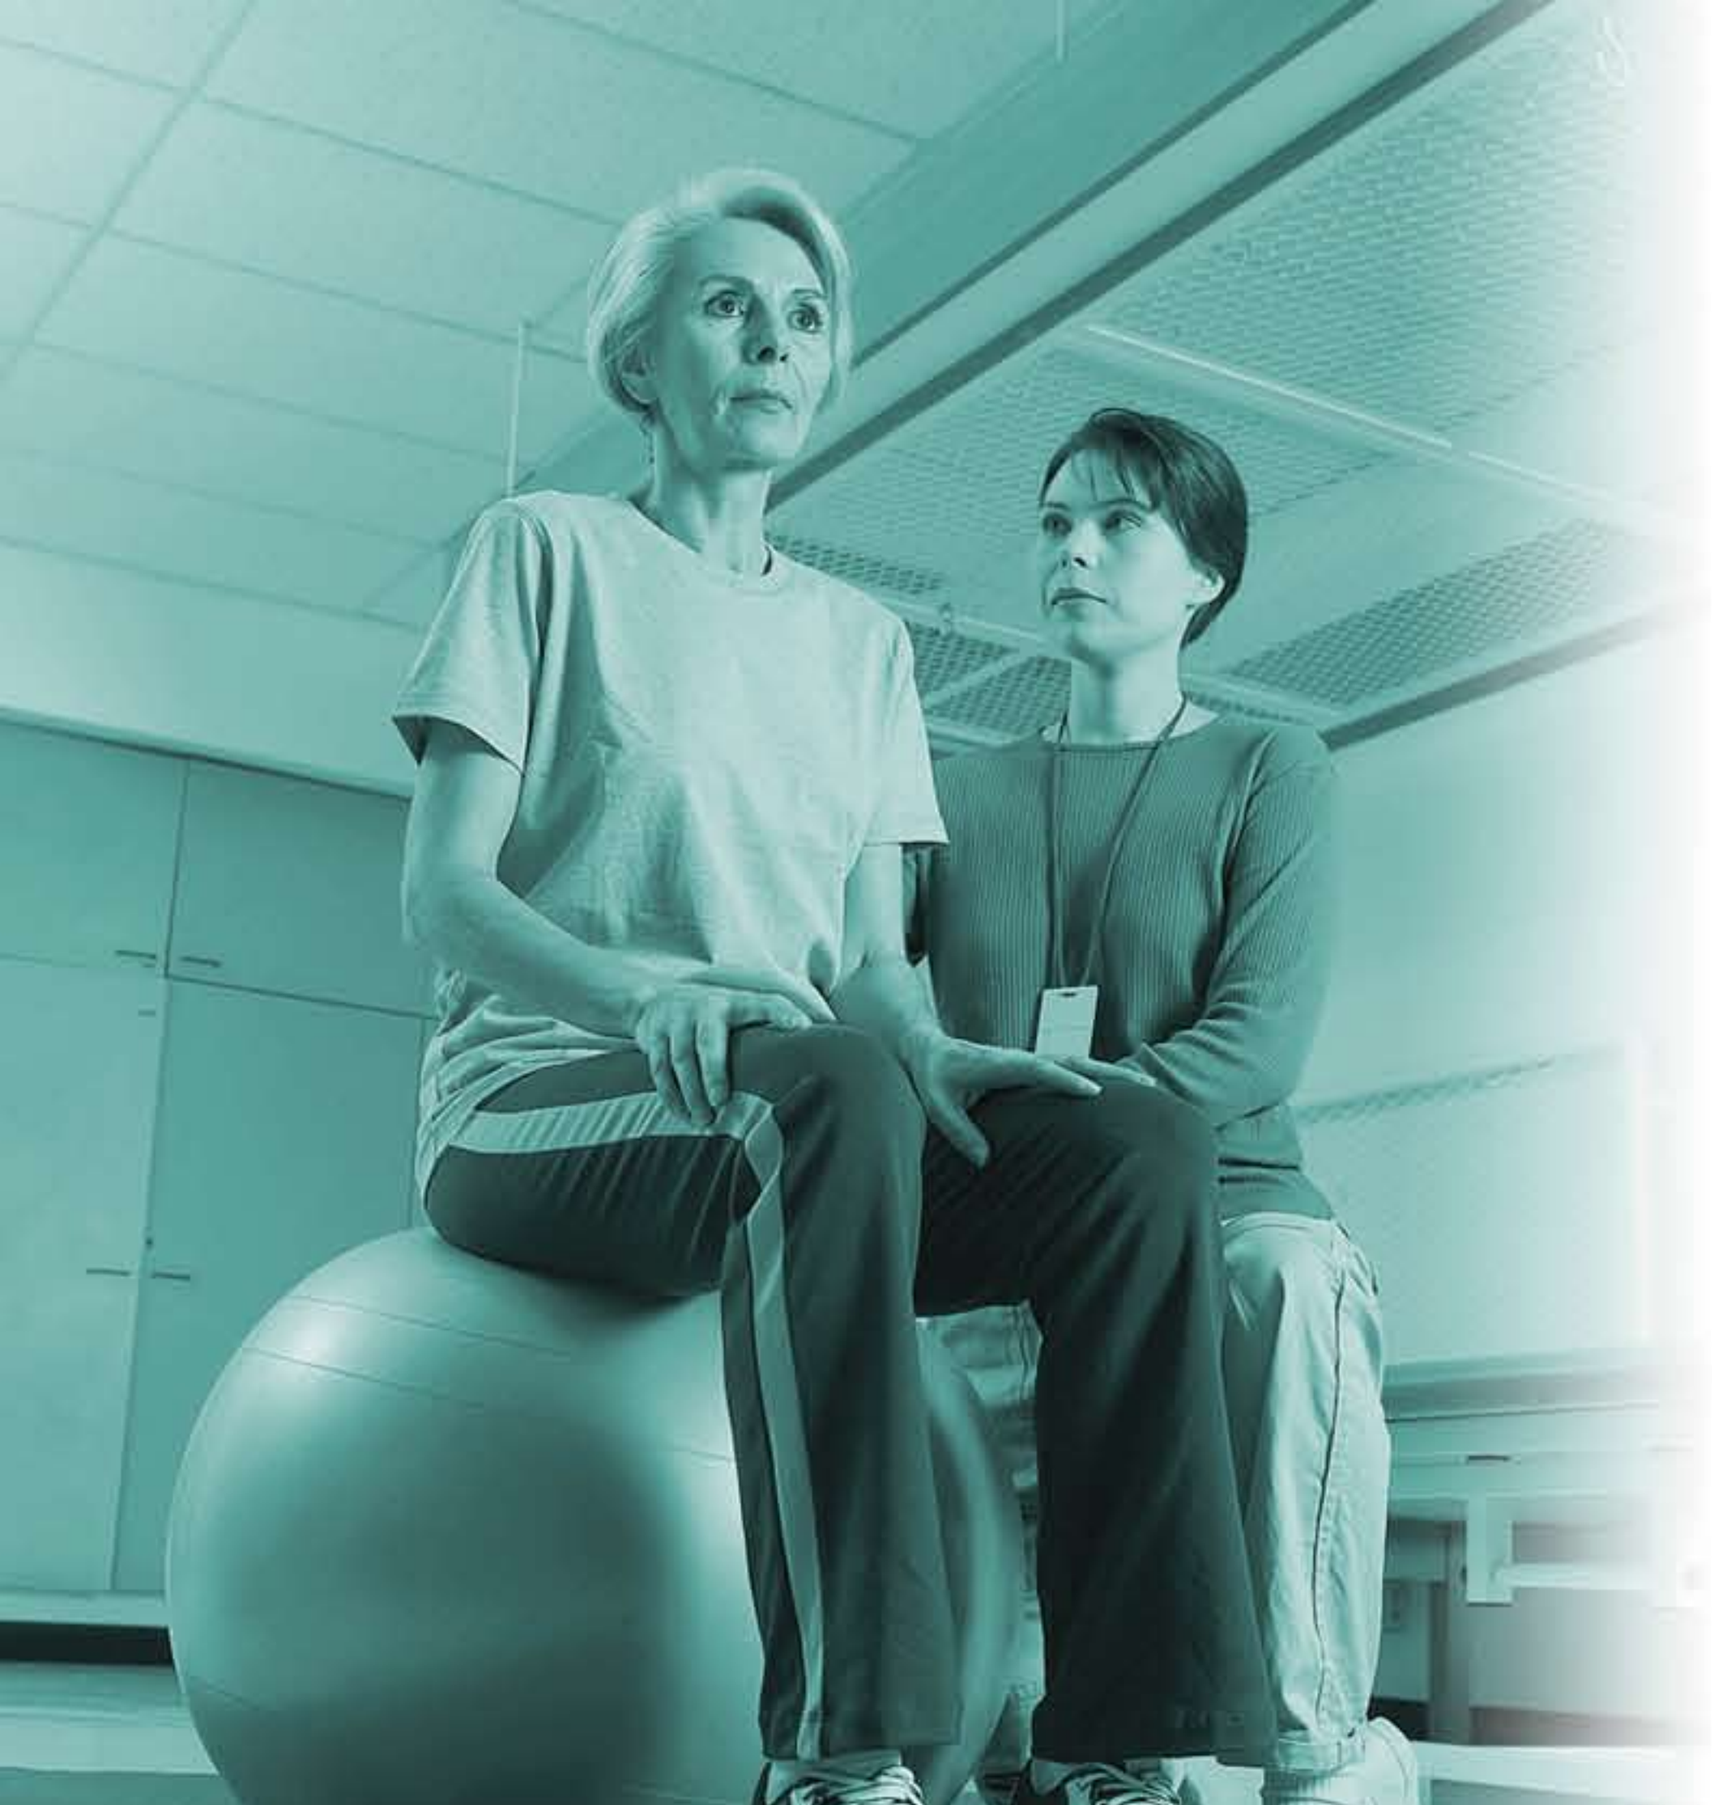

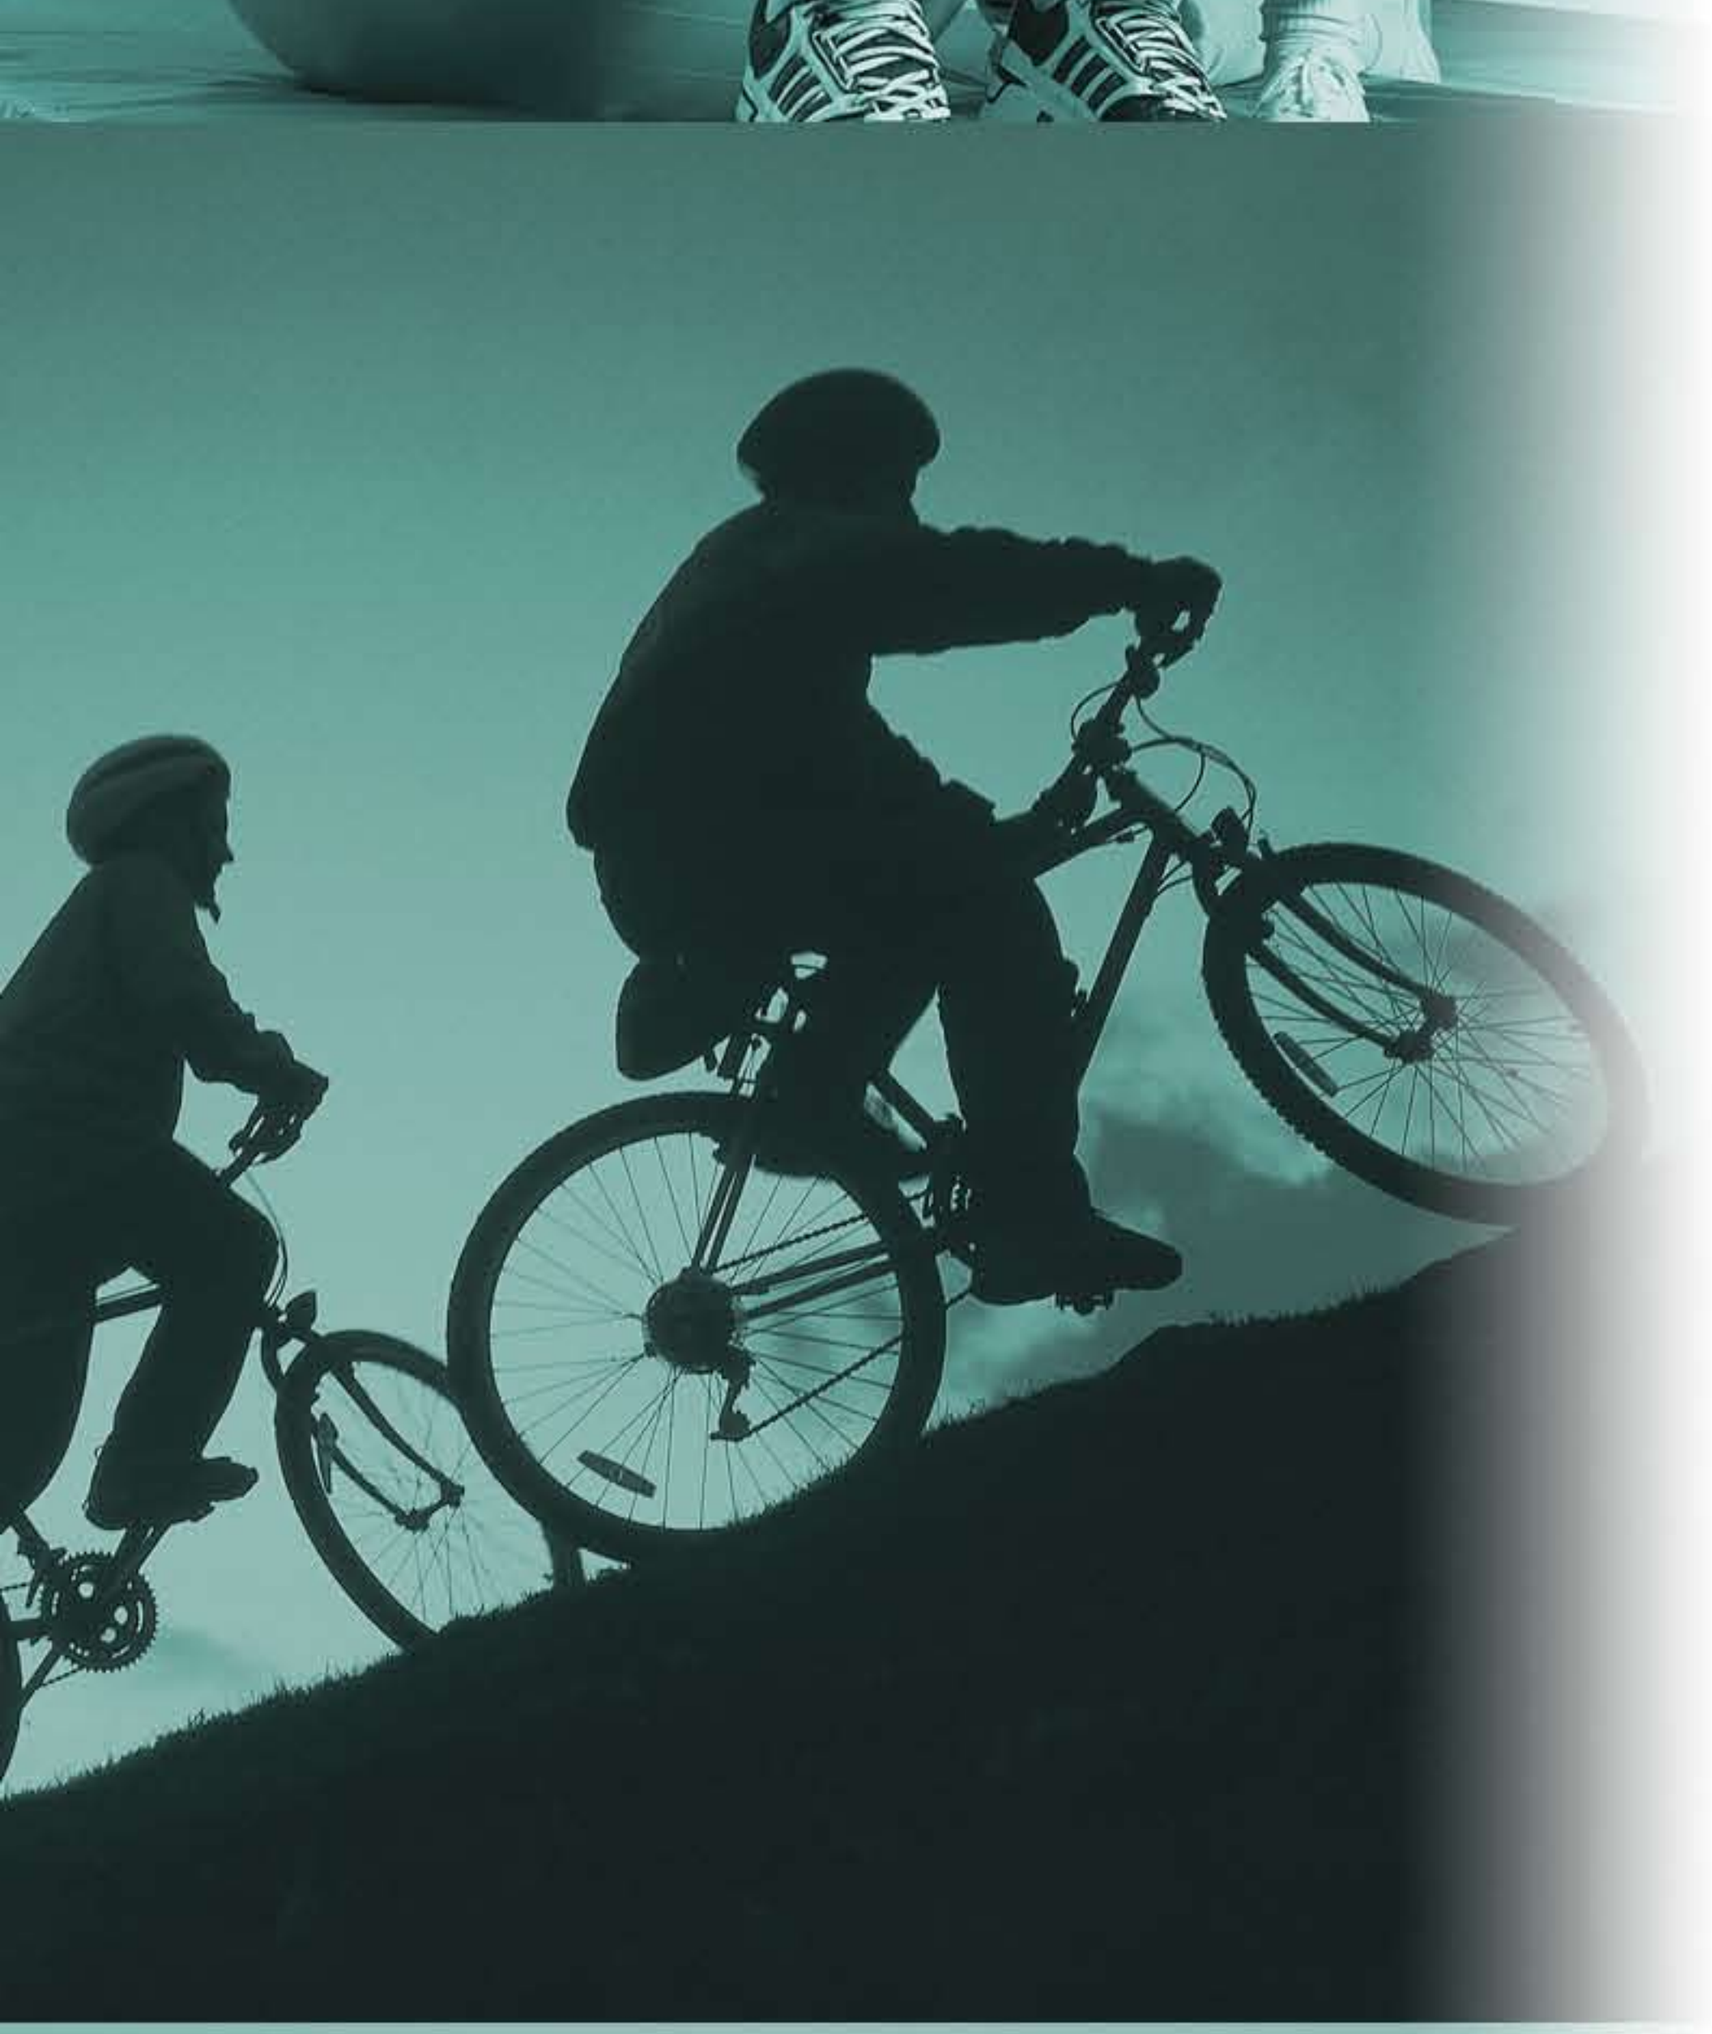

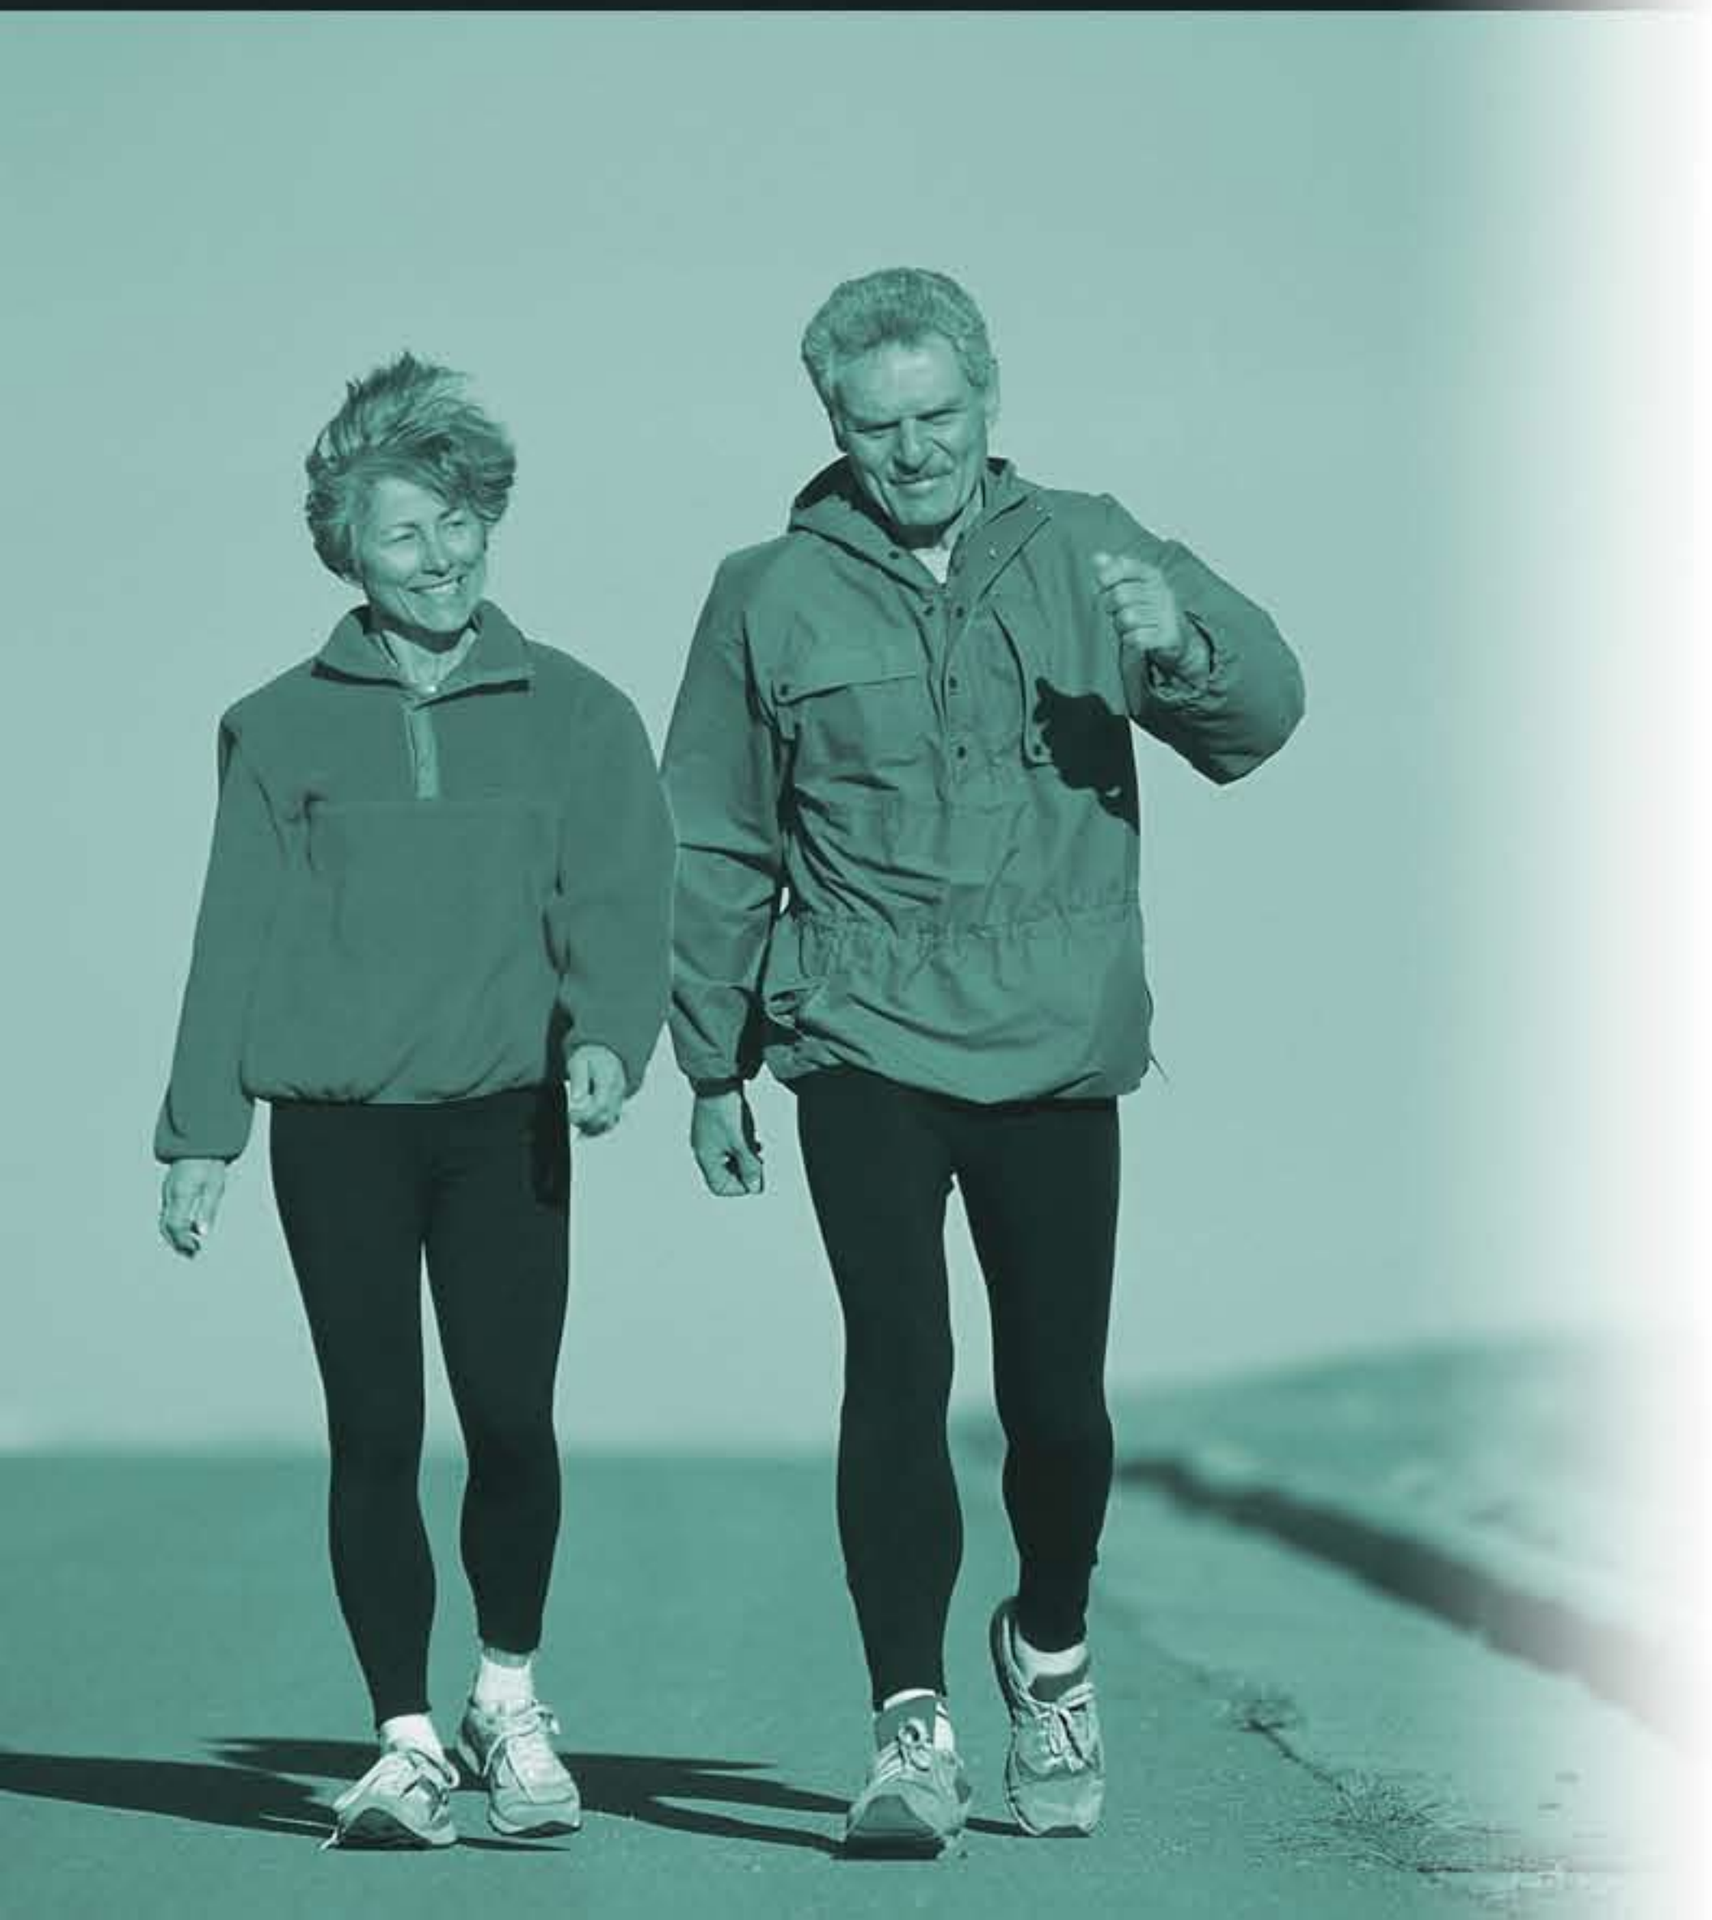

LIPID OPTIMIZATION TOOL (LOT) DATABASE TO ACHIEVE LDL CONTROL IN A COMMUNITY CARDIOLOGY GROUP PRACTICE

Authors: J. Niznick, C. Fulop, V. Savage
Ottawa Cardiovascular Centre (OCC), Ottawa, Ontario, Canada

PURPOSE:

Lipid optimization to newer more aggressive targets has the potential to reduce cardiovascular events significantly. Despite well-established benefits of lipid-lowering therapies, lipid targets are underachieved. At the OCC we have developed a structured physician supervised, nurse managed lipid protocol and applied it via a paper based program. We have audited our practice over the last three years and developed a database capable of ongoing decision support and quality control measurement.

METHODS:

We have previously reported the use of the LOT to guide LDL control in approximately 7000 patients at the OCC. For our 2006 practice audit, a database version of the LOT was developed. This database is designed to risk stratify, to calculate LDL percent reduction to achieve to CCS, ATP III or user defined targets, to provide therapeutic decision support and to track sequential control rates to specified targets. Reports including risk factors, risk modifiers, coronary heard disease (CHD) equivalents and LDL control rates are generated automatically. Parameters from 1002 sequential patients managed with the LOT by 9 OCC physicians were entered into the database.

RESULTS:

Of the 1002 patients audited in 2006, LDL control rates were 92% to an LDL of 3.0 mmol/L, 84% to an LDL of 2.5 mmol/L, 61 % to an LDL of 2.0 mmol/L and 47% to an LDL of 1.8 mmol/L. Eighty % of patients were high or very high risk for cardiovascular events and 87% of these were on a statin. Control rates in statin treated patients were 95% to an LDL of 3.0 mmol/L, 89% to an LDL of 2.5 mmol/L, 66% to an LDL of 2.0 mmol/L and 52% to an LDL of 1.8 mmol/L. Ten percent of patients were on combination therapy with statin + ezetimibe and 5% on combination therapy with statin + fenofibrate. Comparable Vascular Protection (VP)/Guideline Oriented Approach to Lipid Lowering (GOALL) registry control rates were 51%, 22% and 20.8 to LDL's of 2.5, 2.0 and 1.8 mmol/L respectively and Canadian Lipid Study-Observational (CALIPSO) control rates of 64% to an LDL of 2.5 and 19% to an LDL of 1.8 mmol/L.

CONCLUSION:

Although achieved LDL control rates at the OCC utilizing the LOT are among the best reported in the world literature, increased use of combination therapy might result in even better control rates. Further testing of this hypothesis in real time clinical practice using the LOT protocol and database is warranted.

Ottawa Cardiovascular Centre

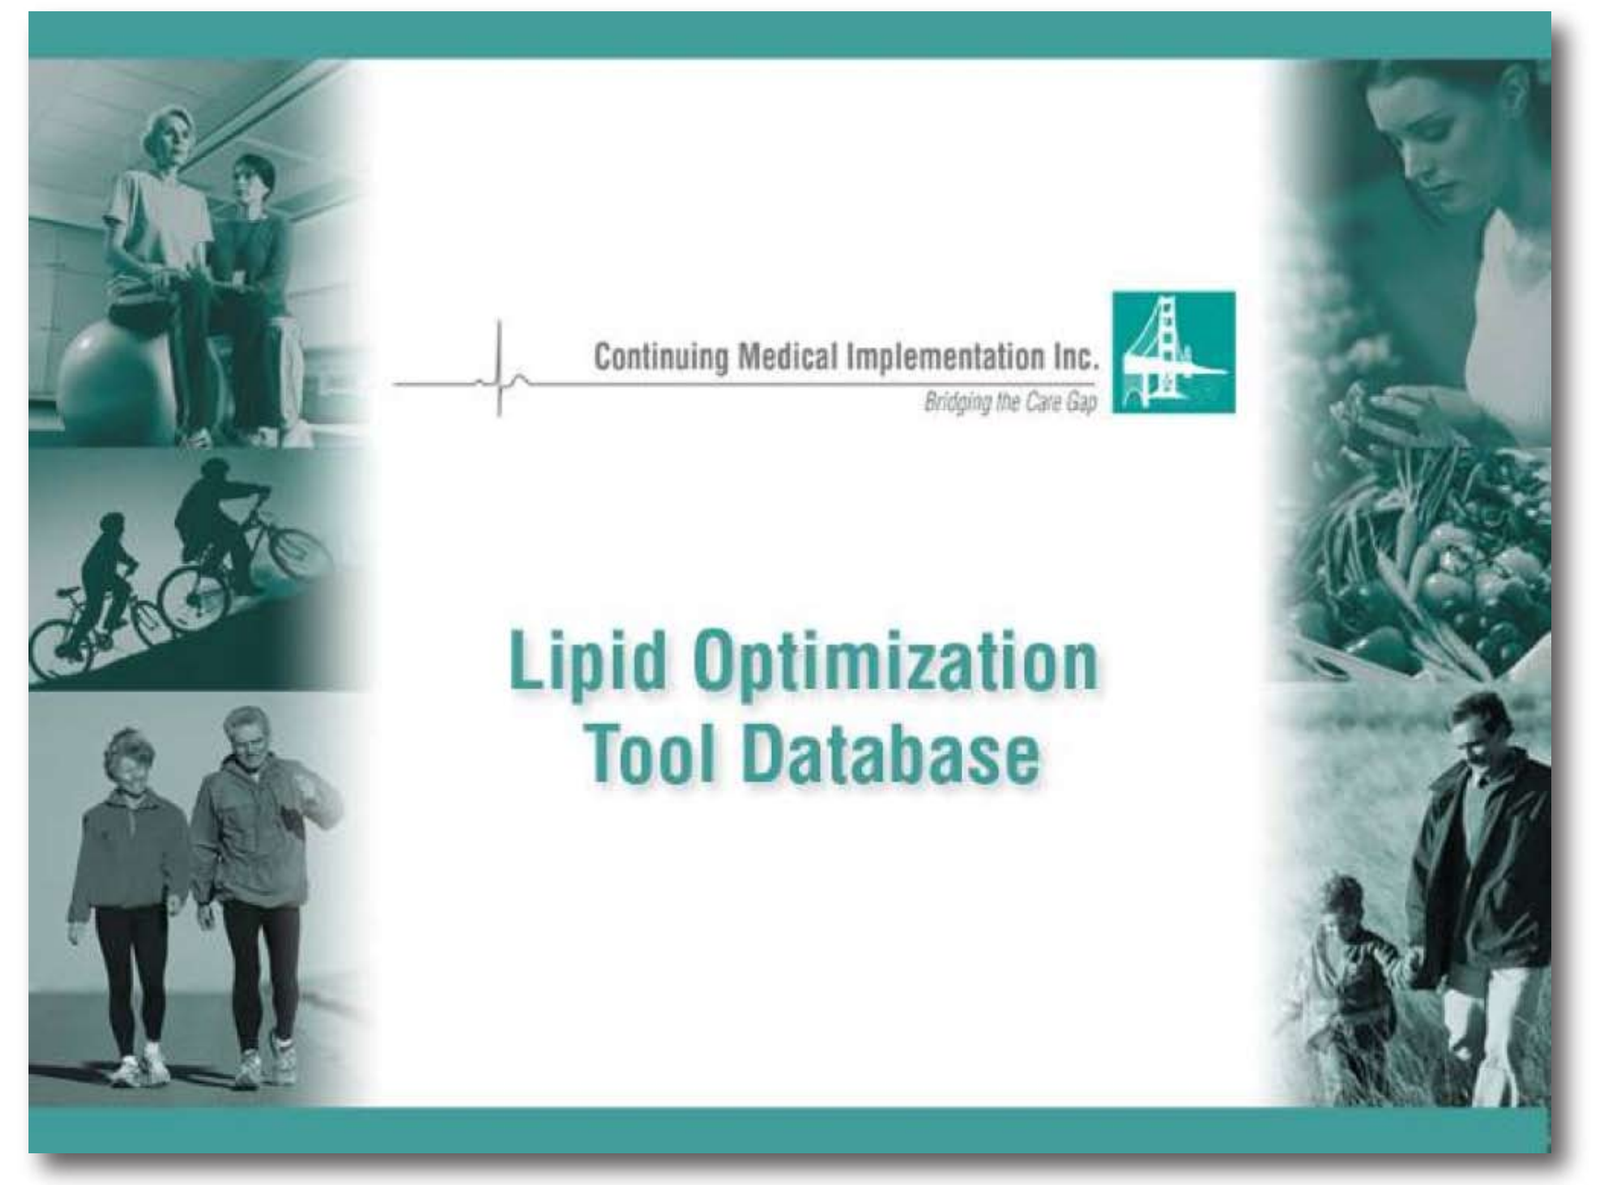
Continuing Medical Implementation Inc.
Bridging the Care Gap
Lipid Optimization Tool Database

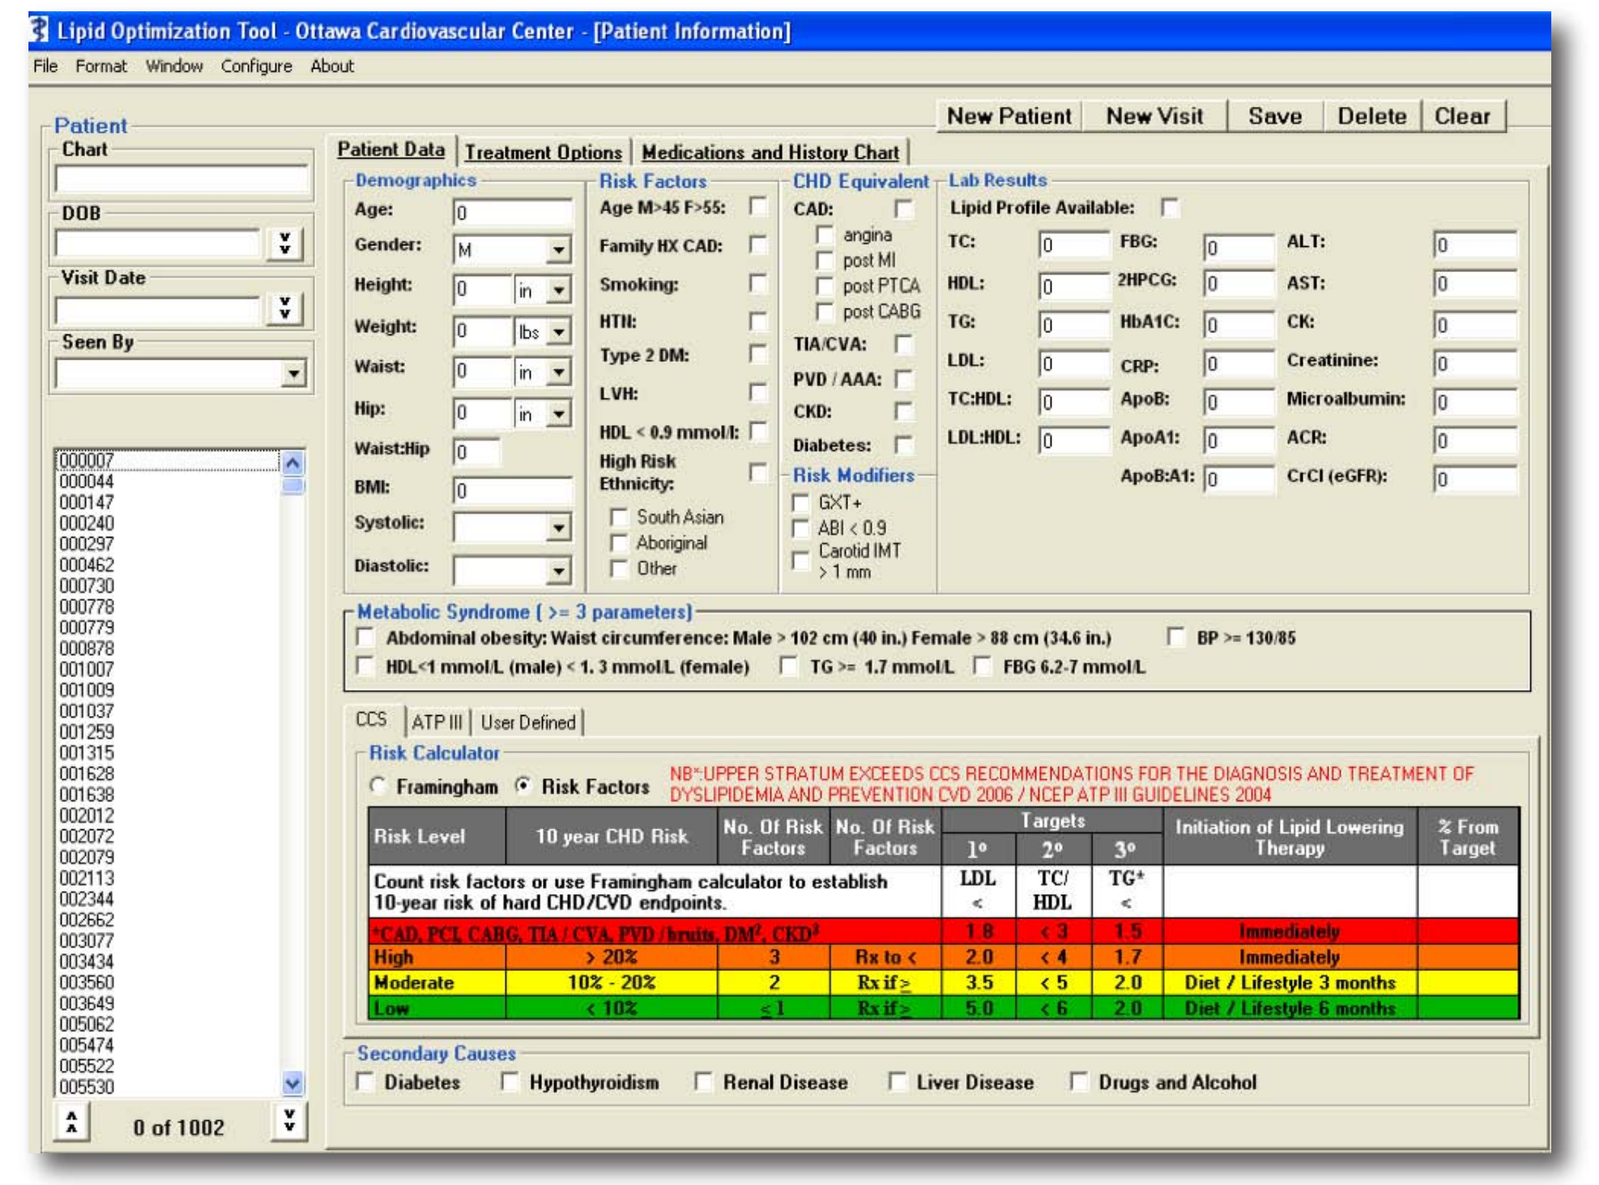
Lipid Optimization Tool - Ottawa Cardiovascular Center - [Patient Information]

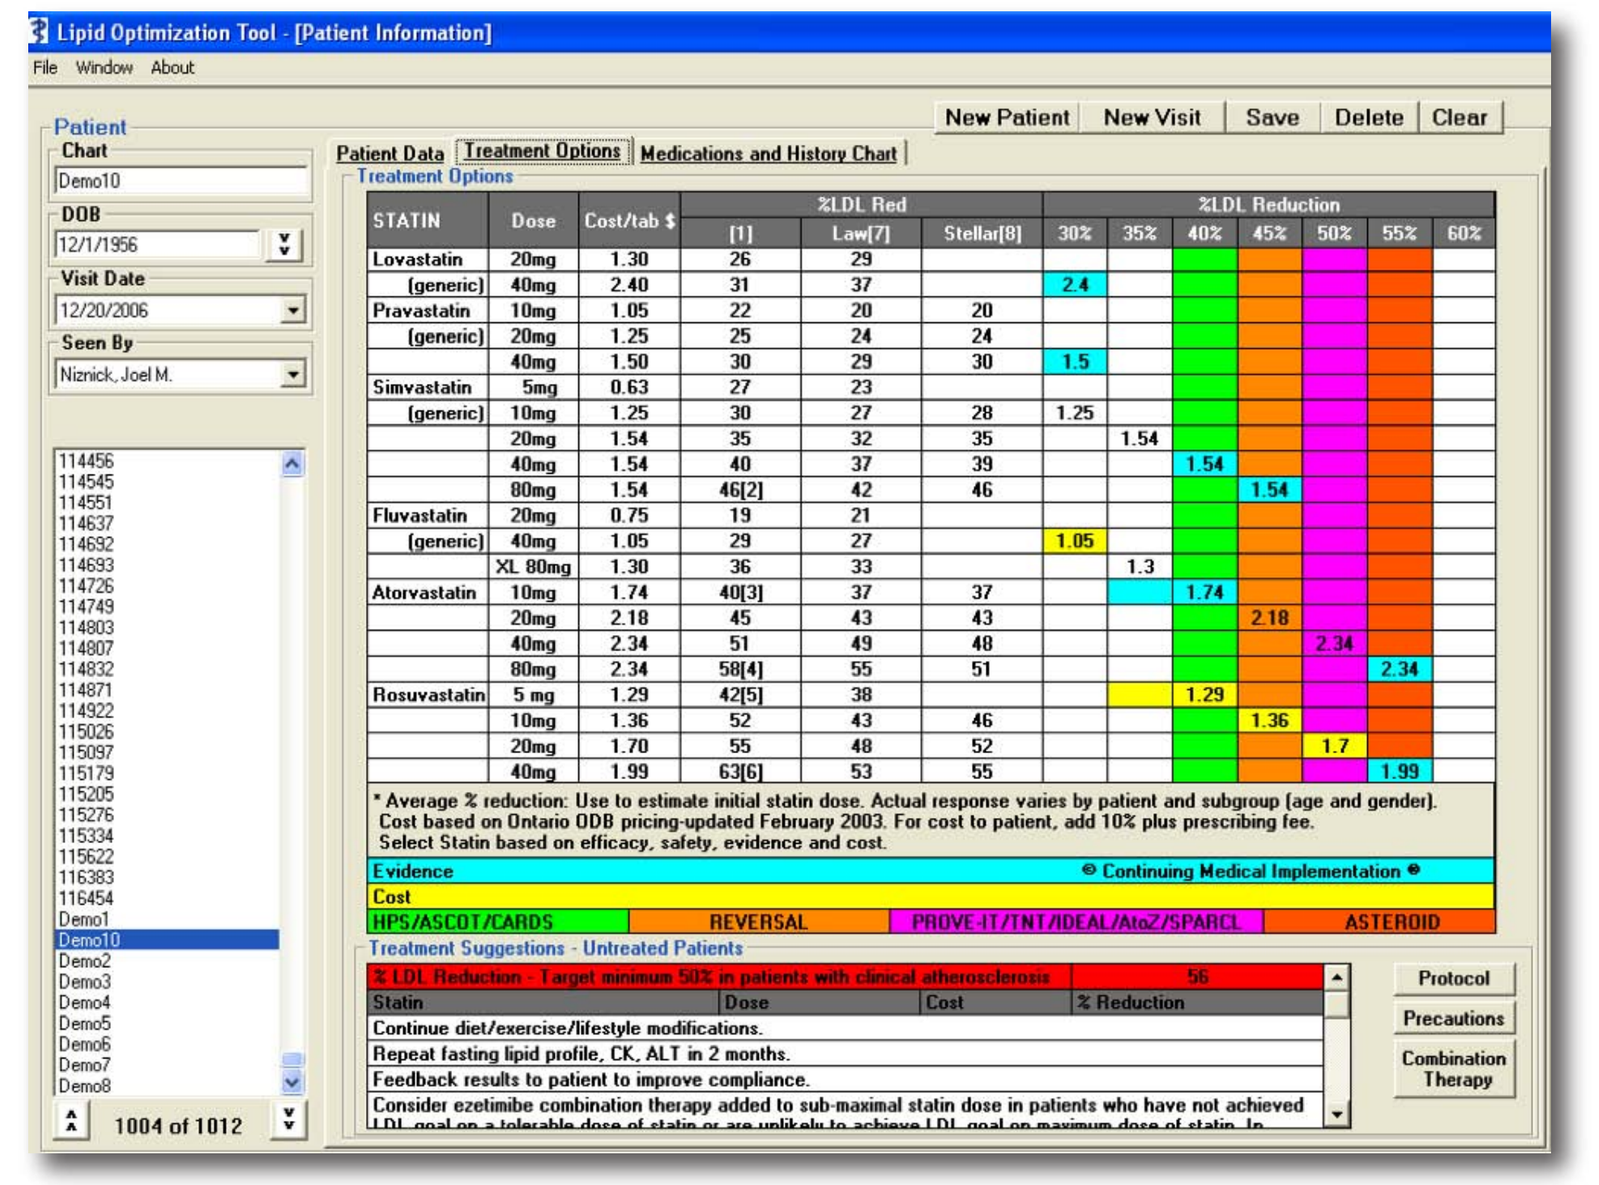
Lipid Optimization Tool - [Patient Information]

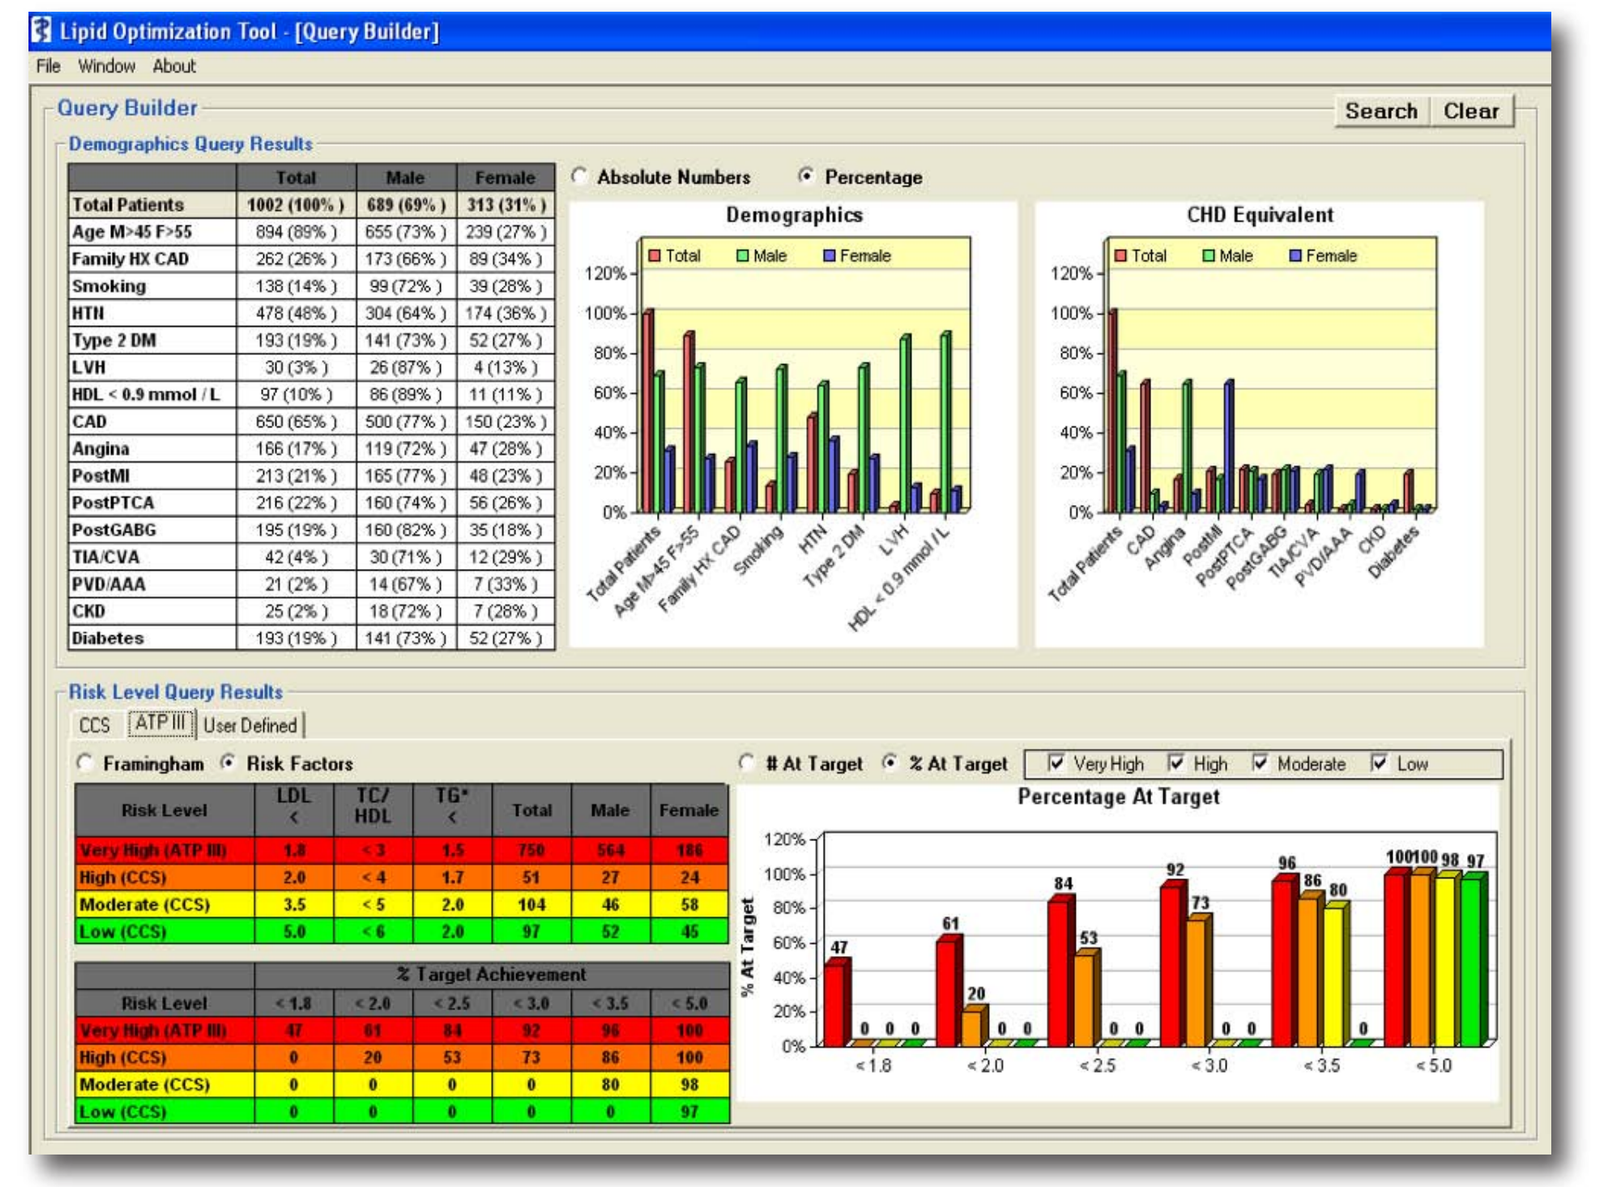
Lipid Optimization Tool - [Query Builder]

Demographic Query Builder Results

	Total	Male	Female
Total Patients	1002 (100%)	689 (69%)	313 (31%)
Age M>45 F>55	894 (89%)	655 (73%)	239 (27%)
Family HX CAD	262 (26%)	173 (66%)	89 (34%)
Smoking	138 (14%)	99 (72%)	39 (28%)
HTN	478 (48%)	304 (64%)	174 (36%)
Type 2 DM	193 (19%)	141 (73%)	52 (27%)
LVH	30 (3%)	26 (87%)	4 (13%)
HDL < 0.9 mmol / L	97 (10%)	86 (89%)	11 (11%)
CAD	650 (65%)	500 (77%)	150 (23%)
Angina	166 (17%)	119 (72%)	47 (28%)
Post MI	213 (21%)	165 (77%)	48 (23%)
Post PTCA	216 (22%)	160 (74%)	56 (26%)
Post CABG	195 (19%)	160 (82%)	35 (18%)
TIA/CVA	42 (4%)	30 (71%)	12 (29%)
PVD/AAA	21 (2%)	14 (67%)	7 (33%)
CKD	25 (2%)	18 (72%)	7 (28%)

OCC LOT Database Audit 2006 ATP III Optional Goal All patients

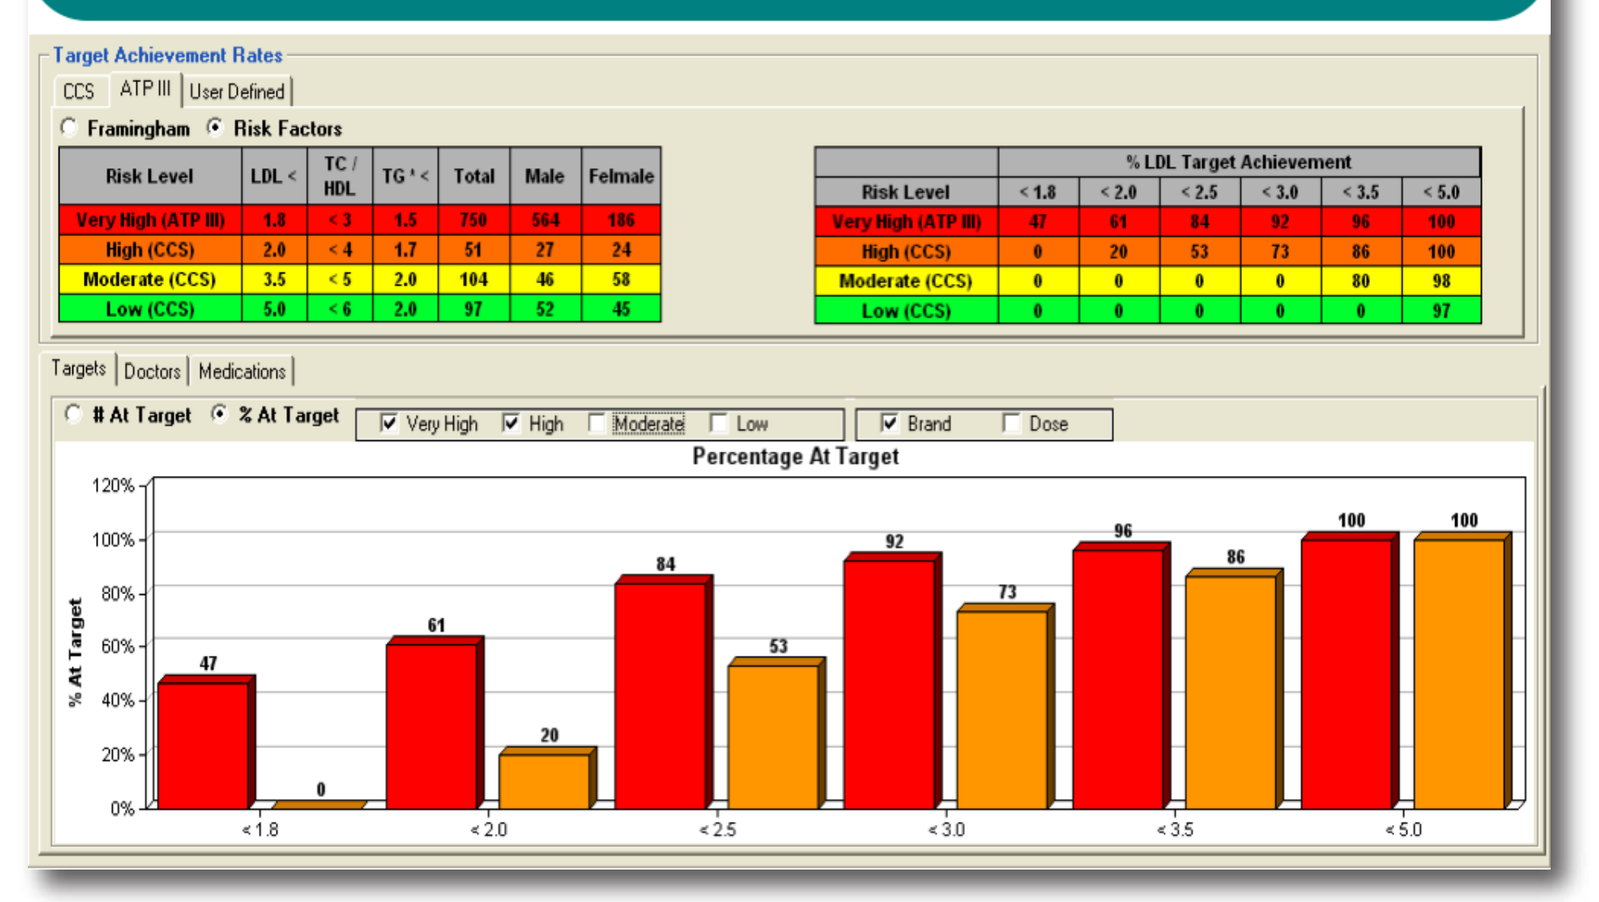
Target Achievement Rates
Percentage At Target

OCC LOT Database Audit 2006 ATP III Optional Goal Patients on Statin

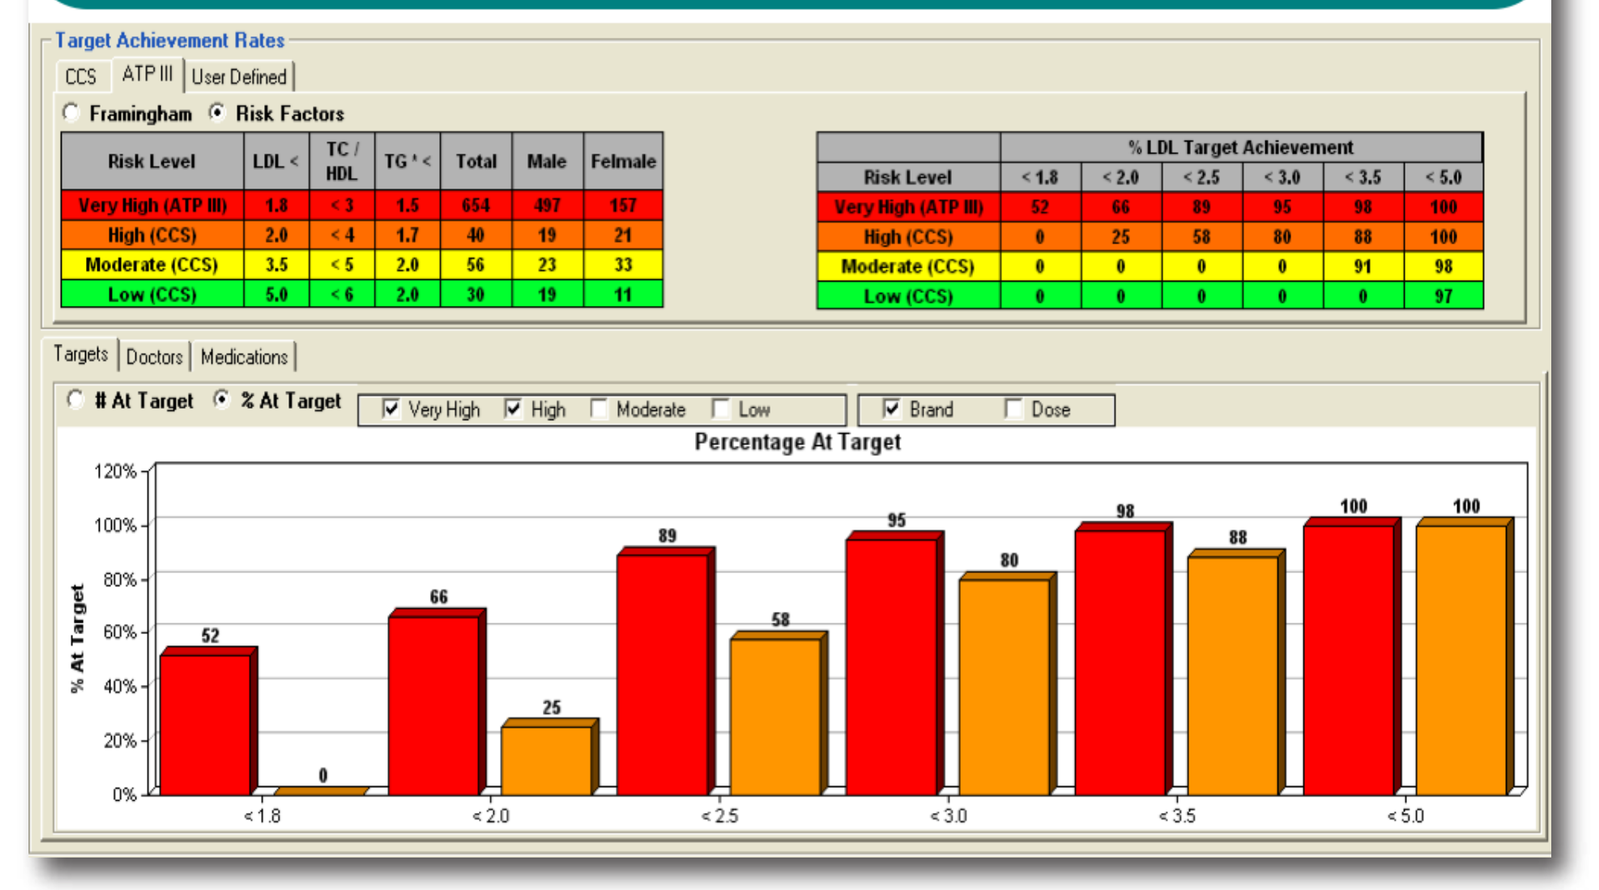
Target Achievement Rates
Percentage At Target

Percent of patients achieving LDL target 2.5 mmol/L (100 mg/dl) in published series

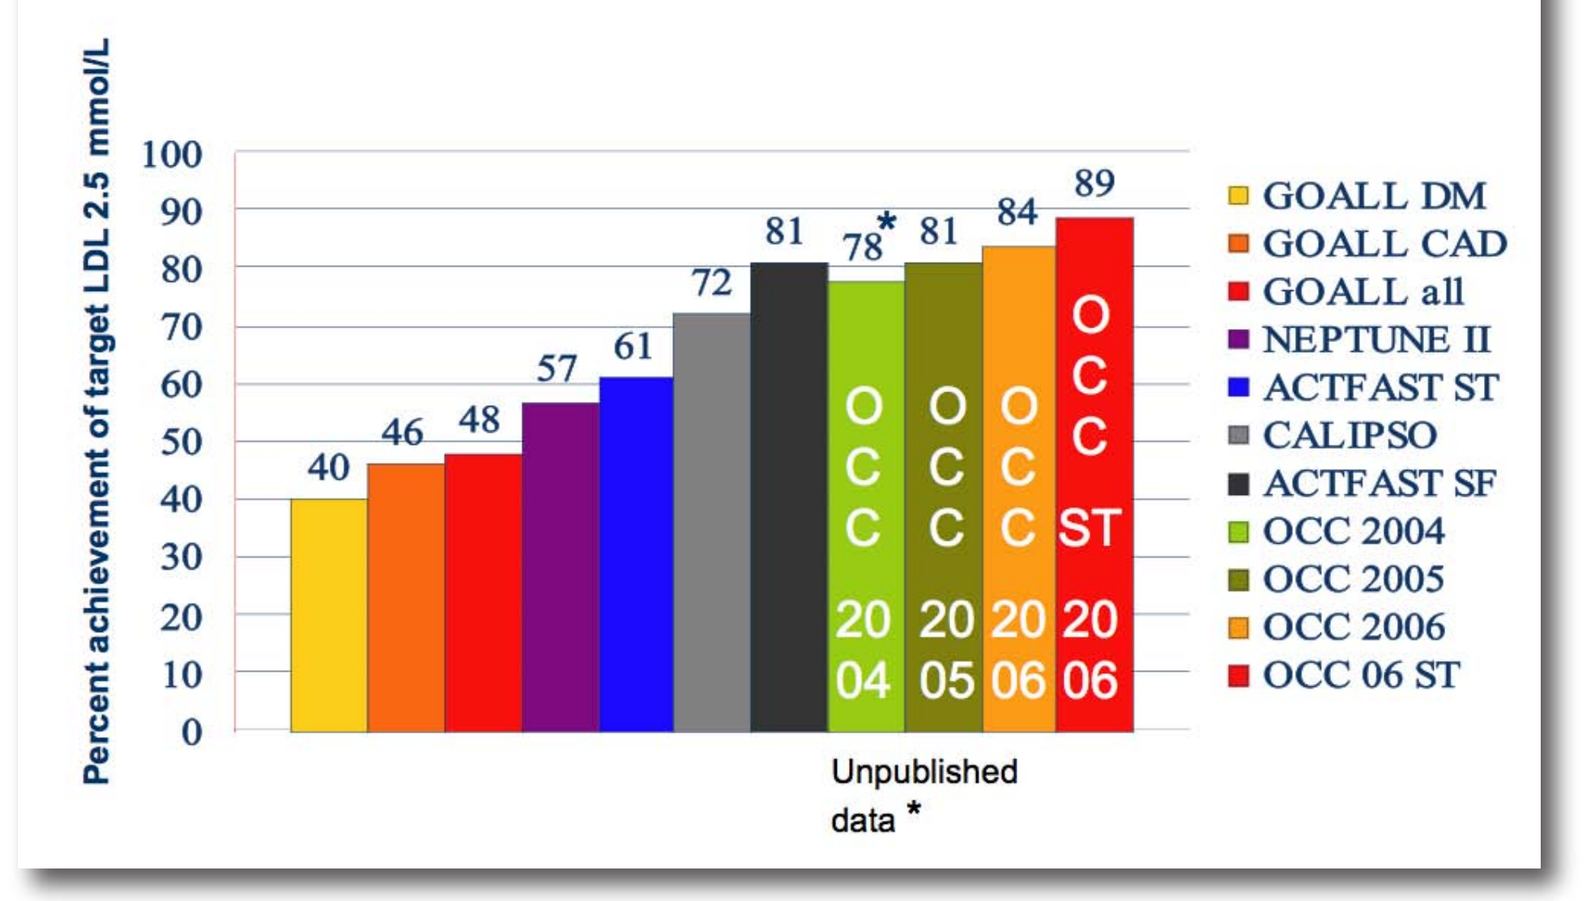
Percent achievement of target LDL 2.5 mmol/L
40
46
48
57
61
72
81
78*
81
84
89
OCC 2004
OCC 2005
OCC 2006
OCC ST 2006
Unpublished data *
GOALL DM
GOALL CAD
GOALL all
NEPTUNE II
ACTFAST ST
CALIPSO
ACTFAST SF
OCC 2004
OCC 2005
OCC 2006
OCC 06 ST

Target LDL Control Rates High and Very High Risk Patients

Target LDL mmol/L	OCC LOT All pts. % at target	OCC LOT on Statin % at target	VP/GOALL % at target	CALIPSO % at target
< 3.0	92	95		
< 2.5	84	89	51	64
< 2.0	62	66	22	
< 1.8	47	52	21	19

1) RT Yan et al. Guideline Oriented Approach in Lipid Lowering (GOALL)Registry data presented at CCC Symposium Oct 2005.
2) MH Davidson et al. Results of the National Cholesterol Education (NCEP) Program Evaluation Project Utilizing Novel e-Technology (NEPTUNE) II Survey and Implications for Treatment Under the Recent NCEP Writing Group Recommendations. Am J Cardiol 2005 Aug 15;96(4):556-63.
3) A Langer et al. Targeted Dosing of Atorvastatin Achieves Cholesterol Targets Quickly in Subjects with Diabetes or the Metabolic Syndrome (The ACTFAST Studies). Can J Cardiol 2005; Vol 21 Suppl C: Abstract 826, 253C.
4) C Bourgault et al. Stain Therapy in Canadian Patients with Hypercholesterolemia: The Canadian Lipid Study – Observational (CALIPSO). Can J Cardiol 2005; 21(13):1187-1193.
5) Yan et al. Contemporary Management of Dyslipidemia in High Risk Patients: Targets Still Not Met. Am J Med 2006; 119: 676-683.

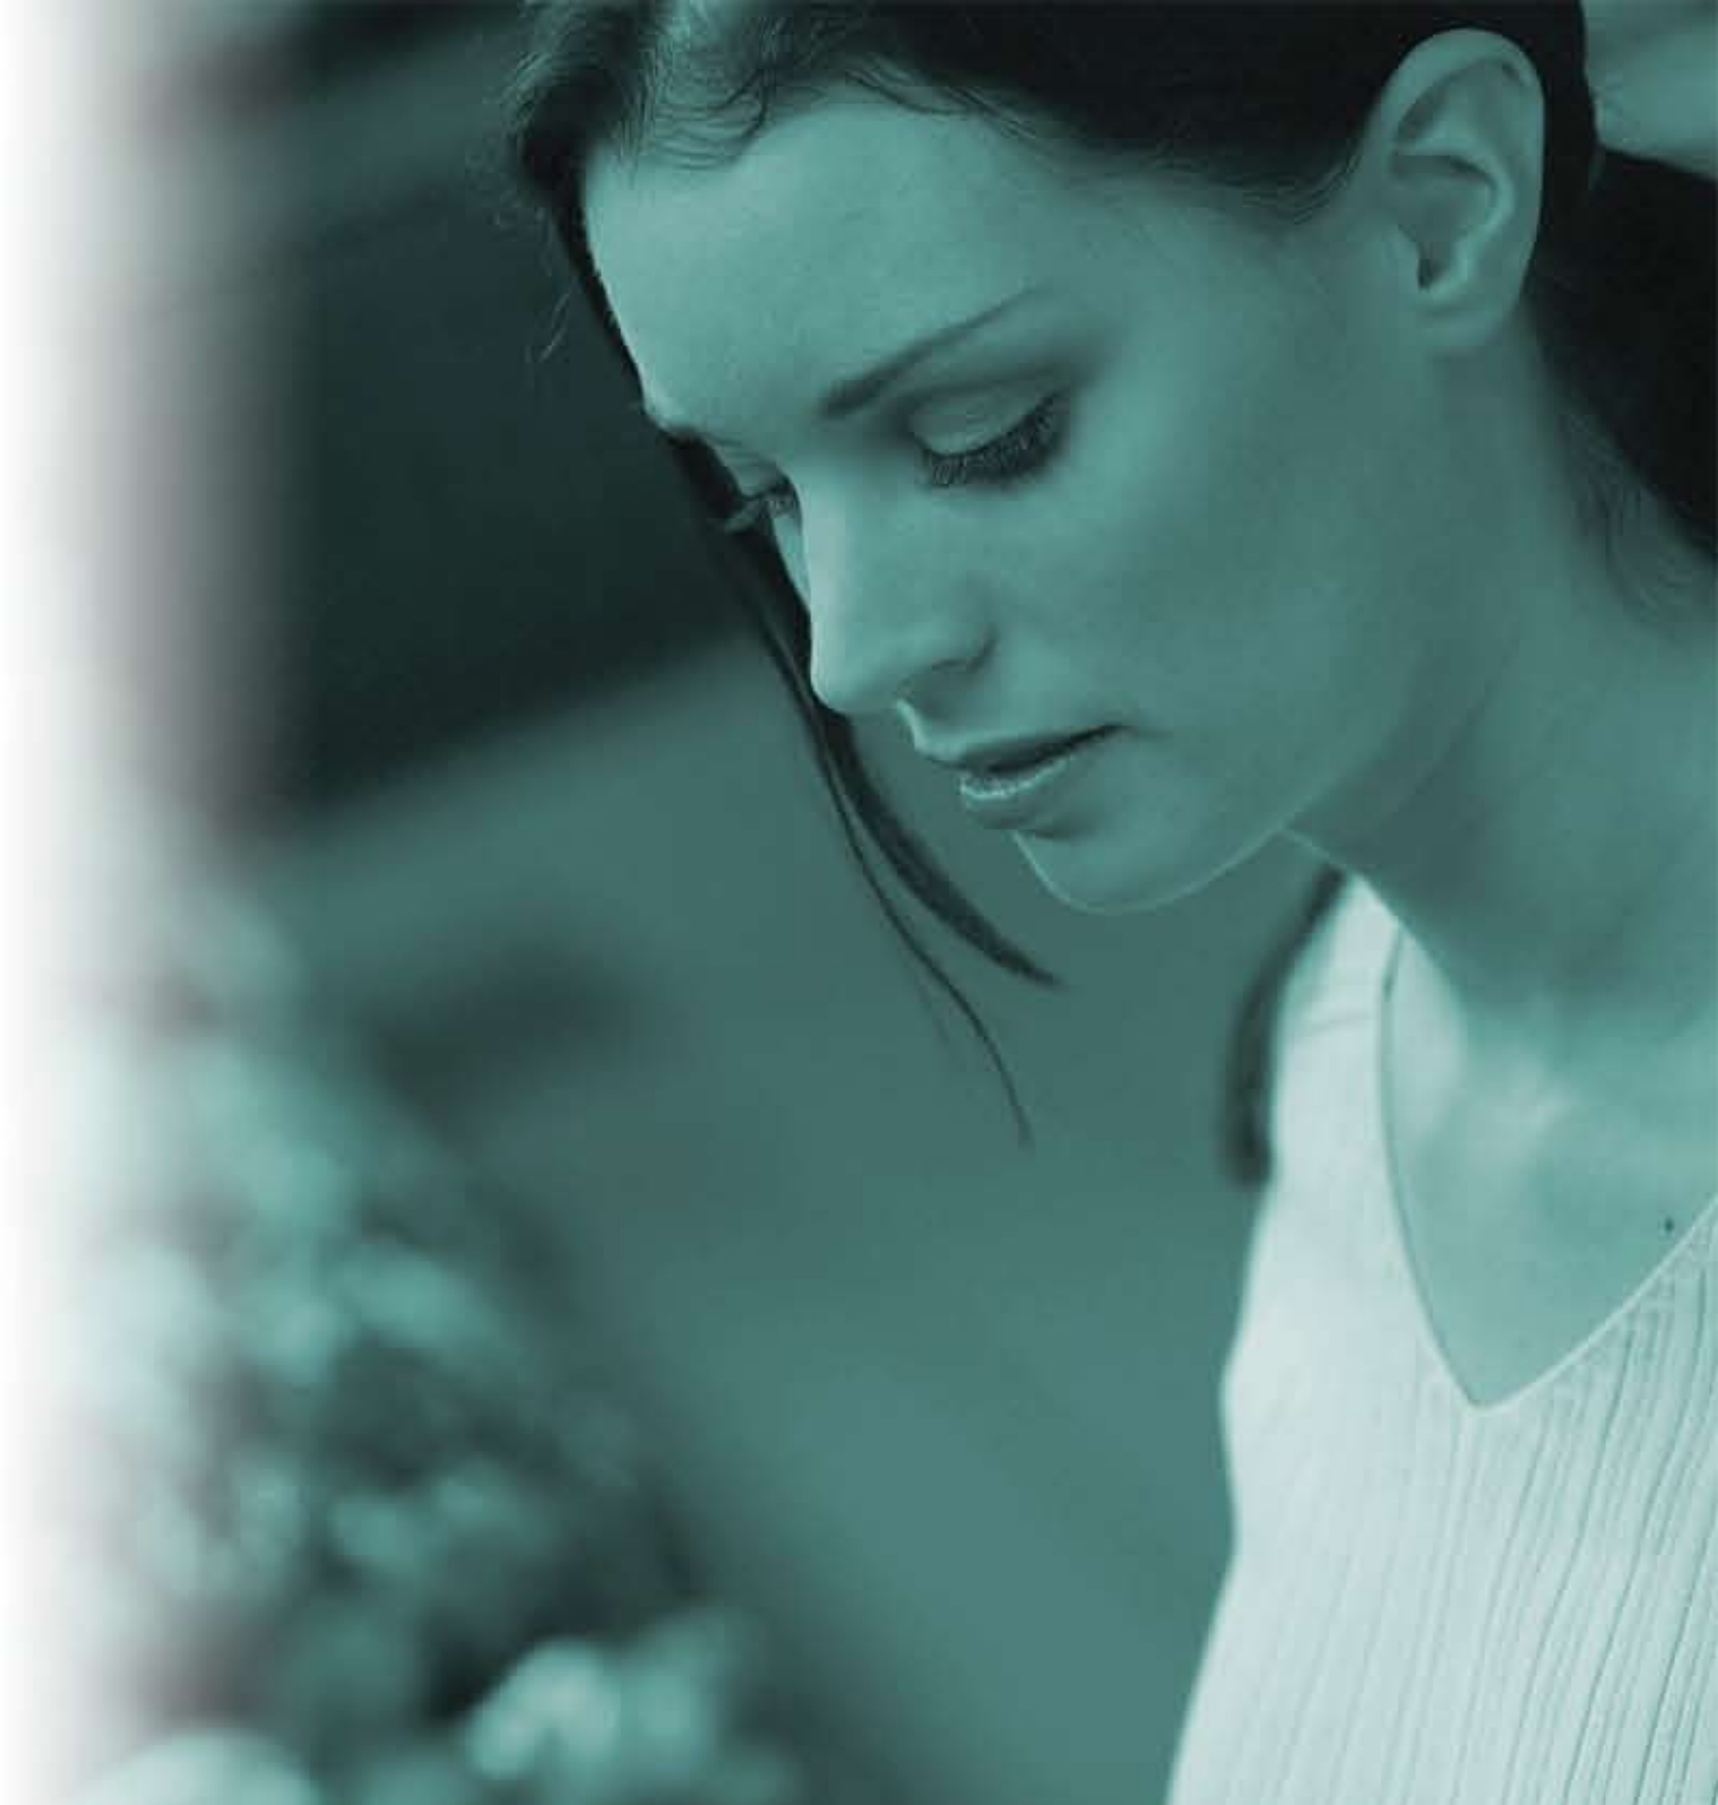

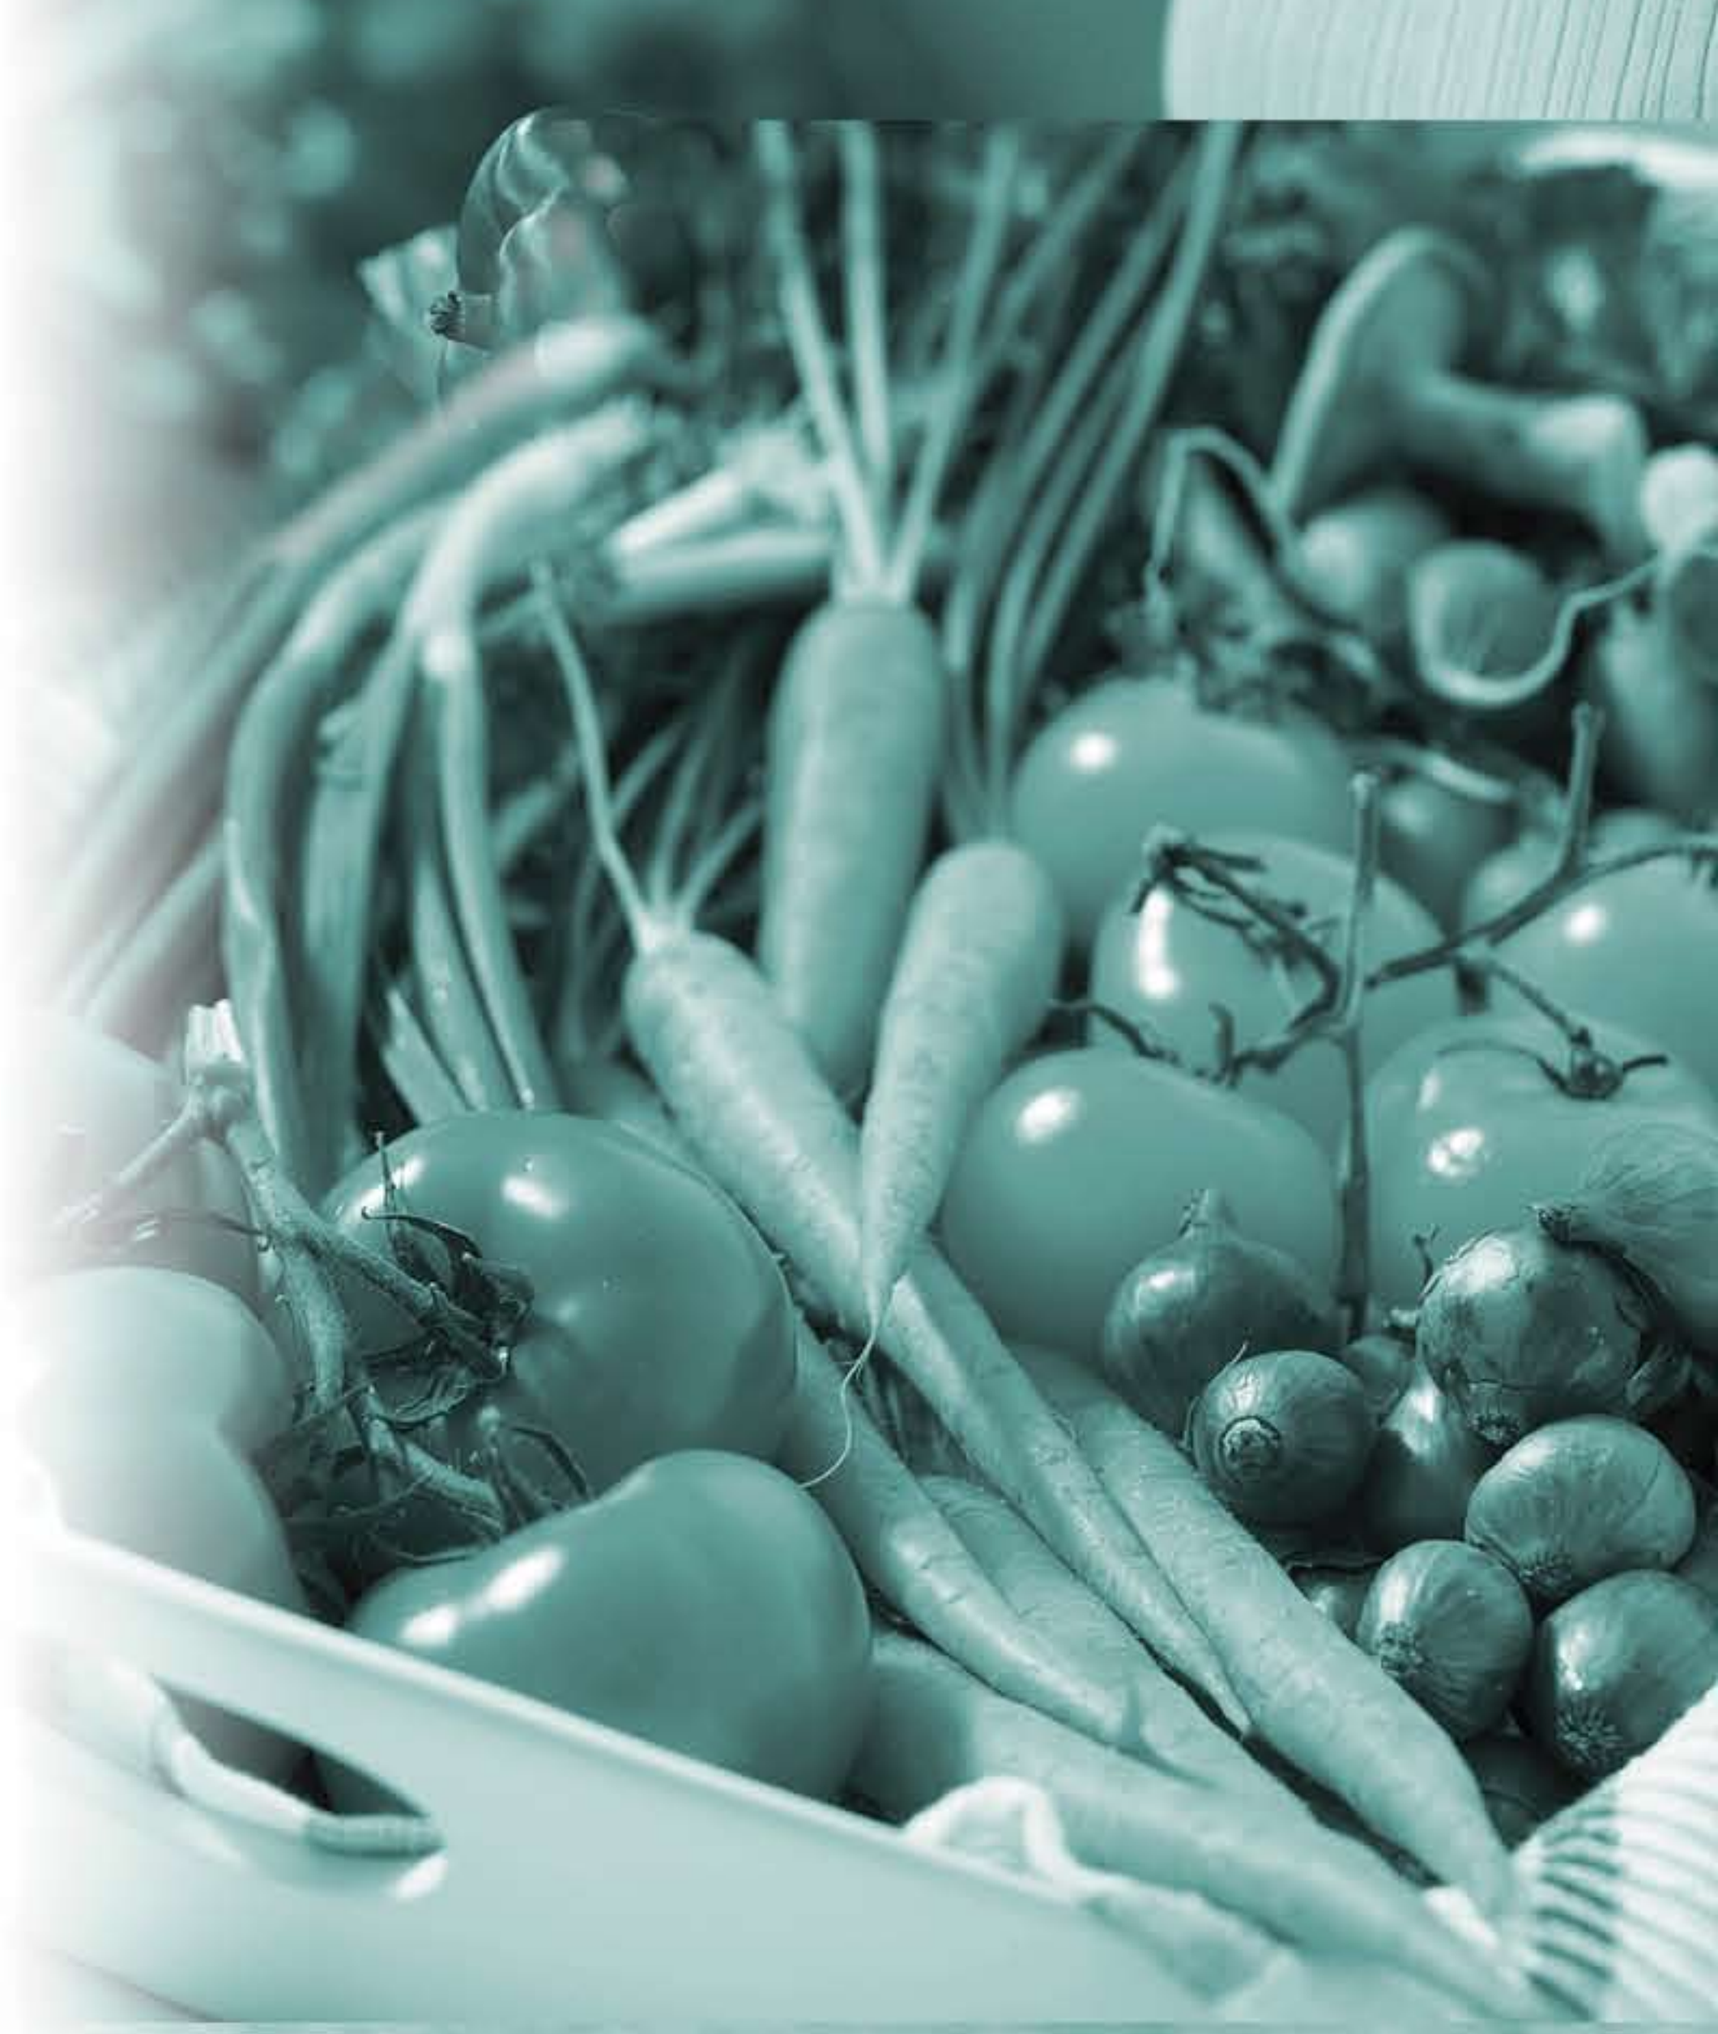

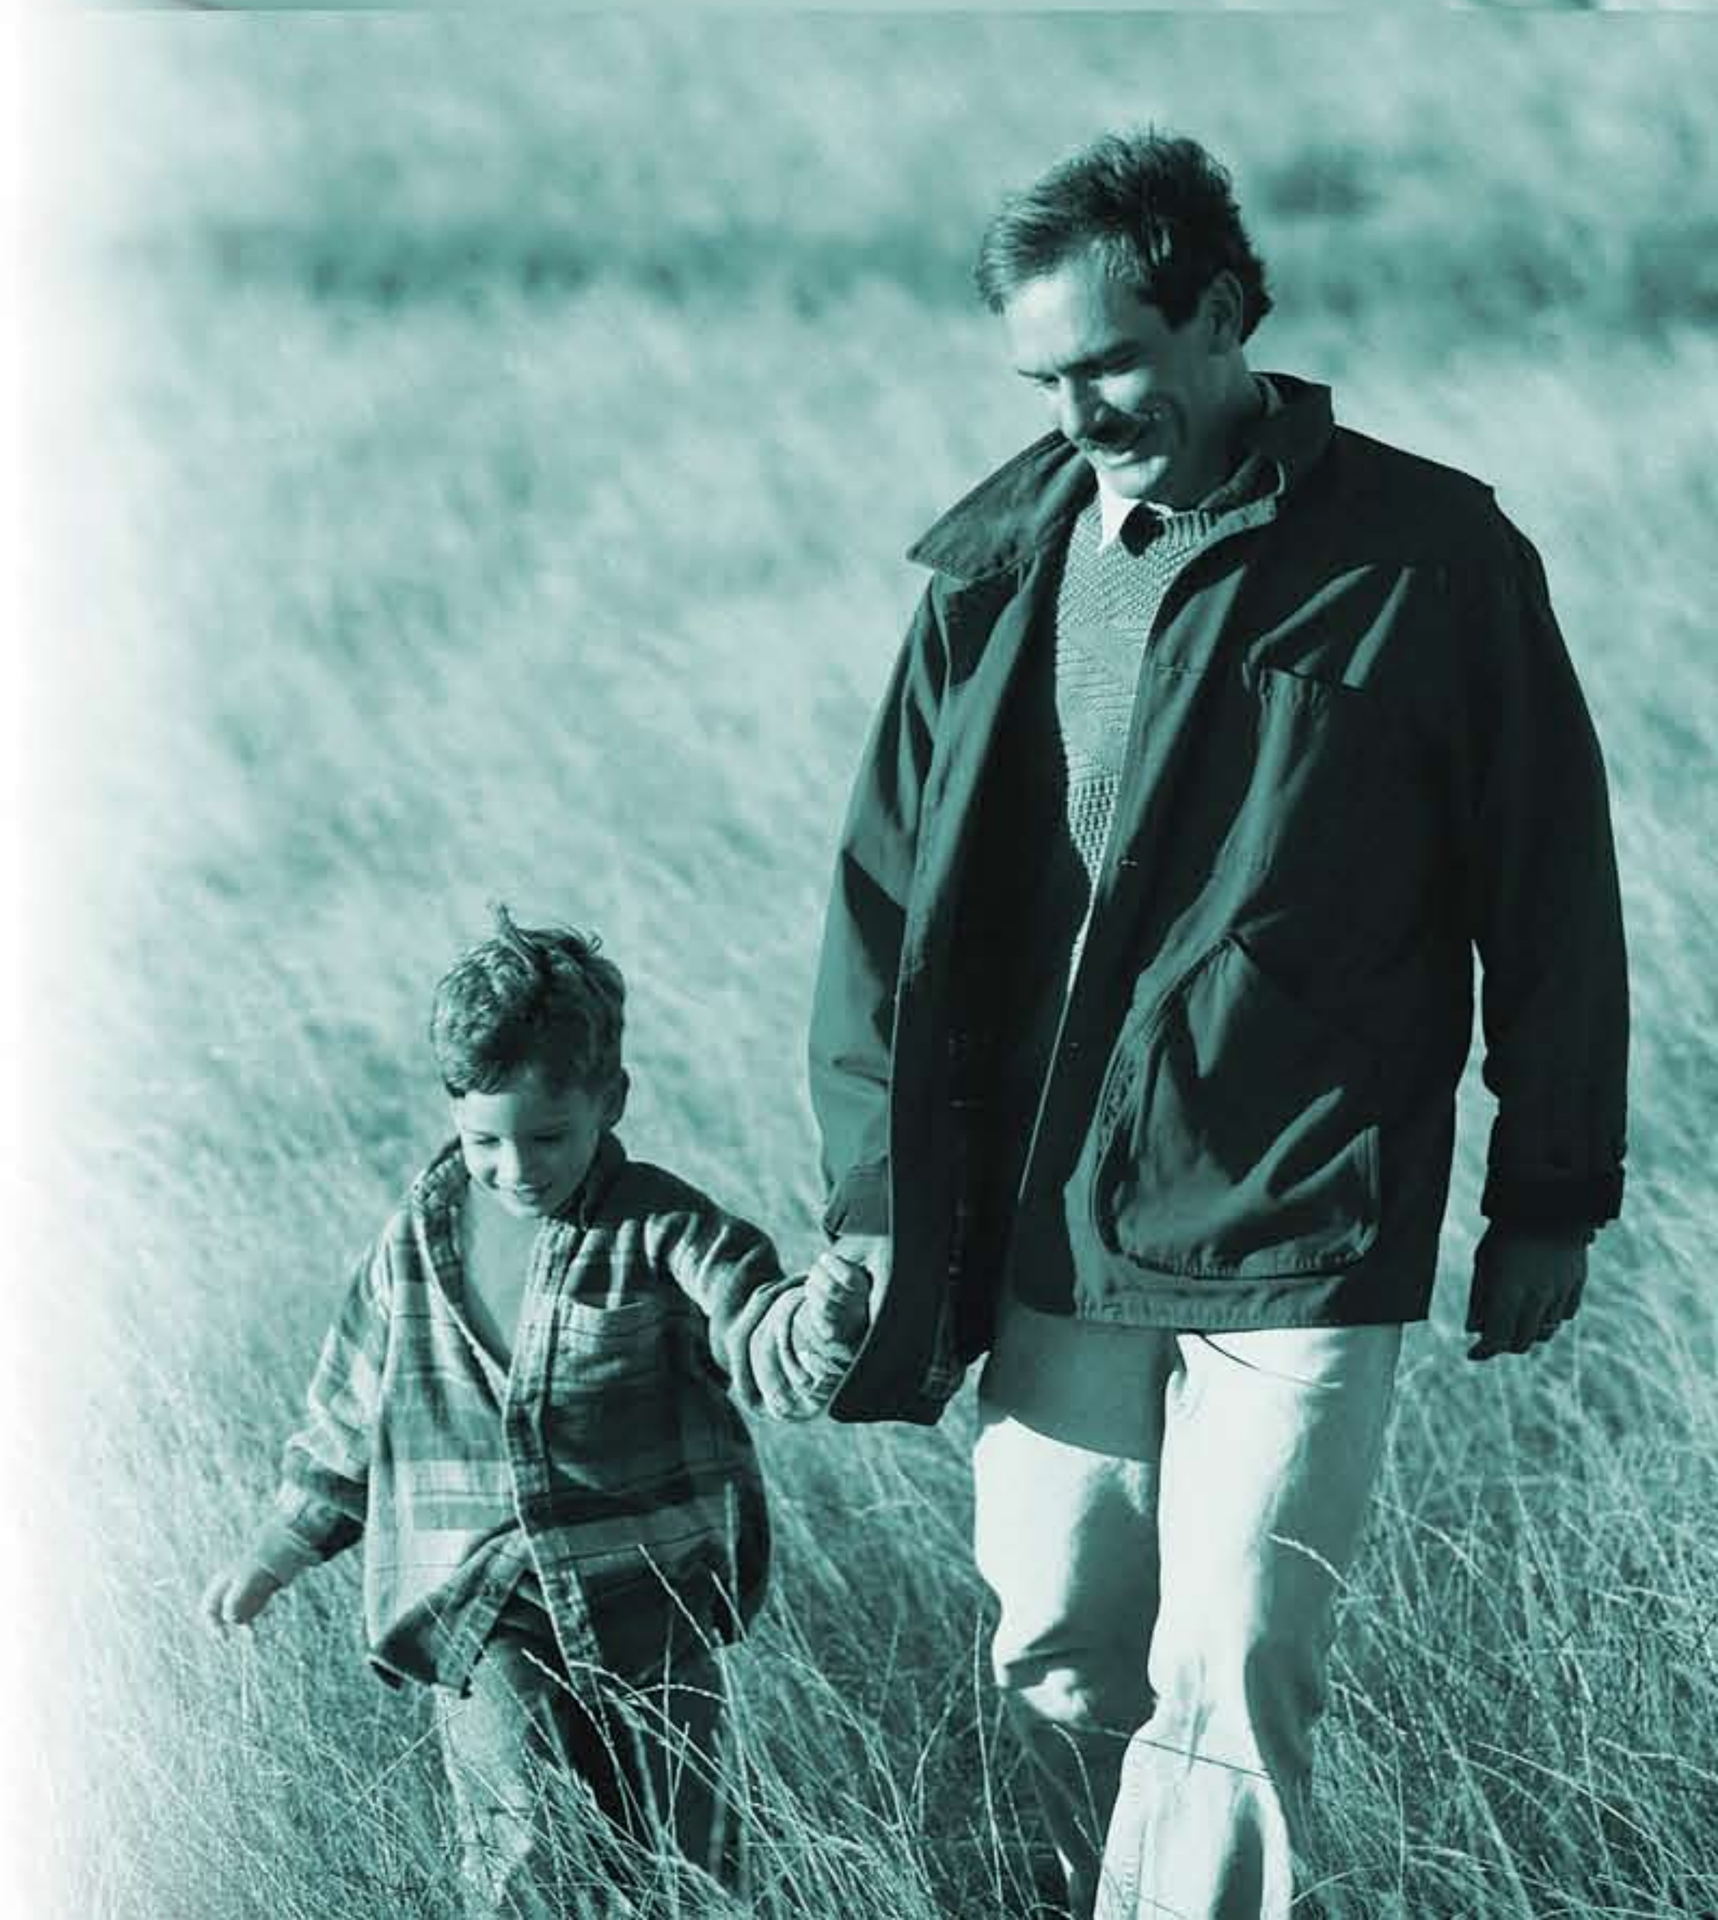